6740

Next-Generation LNPs Induce Effective Anti-Tumor T cell Responses

Douglas G. Millar[2], Matthew Gold[1], Robert Nechanitzky[1], Haritha Menon[1], Yilin Tan[1], Kirstin Olsen[1], Yu Wu[1], Yury Karpov[1], Rajesh Krishnan[1], Pamela S. Ohashi[2], Jae-Kwang Yoo[1], Natalia Martin-Orozco[1]

[1]Providence Therapeutics Holdings, Inc., 8832 Blackfoot Trail SE Calgary, AB, T2J 0T2, [2]Princess Margaret Cancer Centre, 610 University Ave. Toronto, ON, M5G 2M9

INTRODUCTION

Recently, LNP-based mRNA vaccines have gained widespread attention and offer an attractive modality for personalized cancer therapies. However, inducing an effective anti-tumor response often requires the induction of a T cell response breaking self-tolerance mechanisms, a challenge for current therapies. Here, we developed new lipid nanoparticle (LNP) formulations which can be utilized for therapeutic mRNA/LNP cancer vaccines. Utilizing an *in vivo* screening model identifying LNP formulations capable of breaking self-tolerance allows the selection of LNP candidates with superior anti-tumor efficacy. In the well characterized mouse syngeneic colorectal cancer (CRC) model expressing model glycoprotein antigens from lymphocytic choriomeningitis virus (LCMV, MC38gp), intramuscular administration of this new LNP formulation as a monotherapy in a therapeutic setting after tumor inoculation significantly delayed tumor growth and cleared tumors in 50% of treated mice. The data suggests the possible applicability of our next generation LNP formulations for the development of effective therapeutic mRNA cancer vaccines for multiple solid tumors.

Methodology

1. *In vivo* Immunogenicity – mRNA encoding 3 immunodominant epitopes to LCMV (3Gp: gp33, gp61 and gp276) was formulated into LNPs to screen (LNP-01 & LNP-02) and delivered to C57BL/6 animals to evaluate induction of antigen specific CD8$^+$ T cells
2. Breaking self-tolerance – Successful LNP candidates were tested in the RIPgp model of autoimmune diabetes. These transgenic mice express LCMV under control of the rat insulin promoter resulting in gp expression in pancreatic islet β cells. Driving an immune response against this "self"-tissue can be monitored via peripheral blood glucose levels. This provides a robust assay for LNPs capable of inducing a T cell response capable of breaking self-tolerance against a target tissue
3. Tumor control – LNPs displaying a positive result in RIPgp transgenic mice were further tested in a therapeutic setting as a monotherapy for the colon adenocarcinoma model (MC38gp) to assay potential for cancer therapeutics

Immunization with next-generation LNP induces antigen-specific CD8$^+$ T cell responses *in vivo*

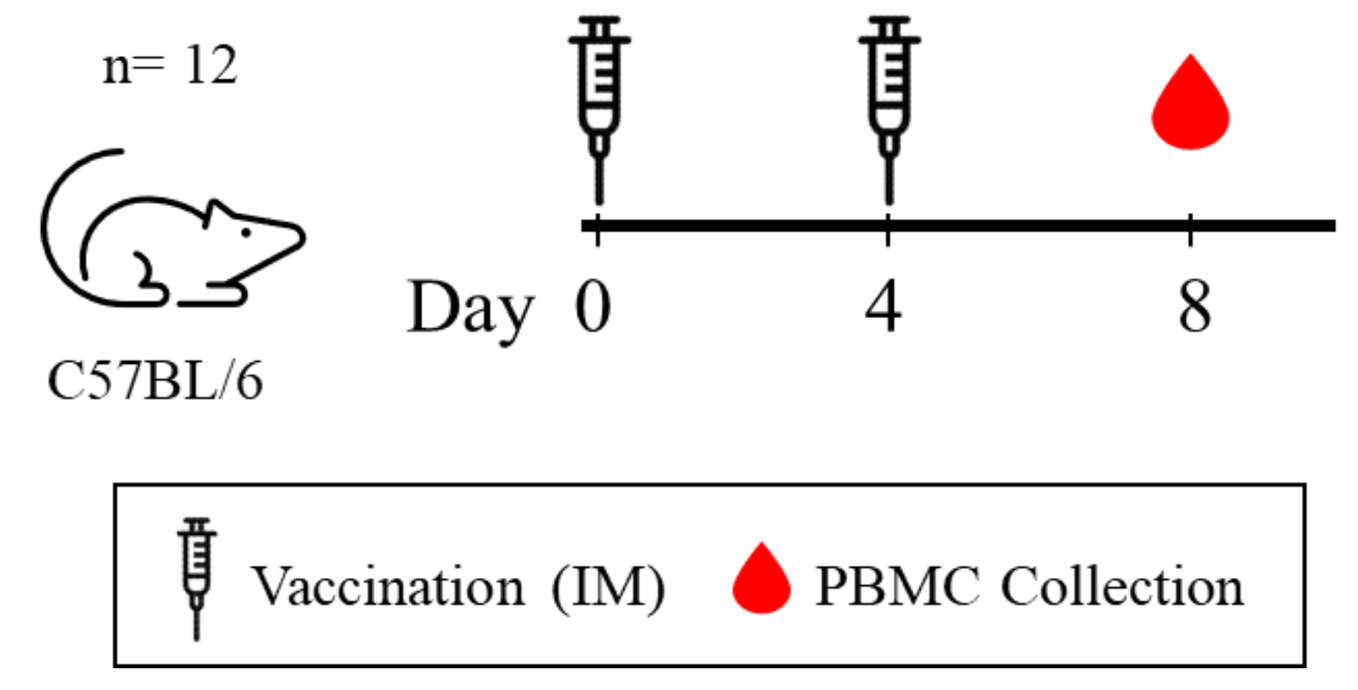
n= 12
C57BL/6
Day 0 4 8
Vaccination (IM)
PBMC Collection

- Both LNP-01 and LNP-02 induce a robust expansion of antigen-specific CD8$^+$ T cells
- BMDCs preferentially prime gp34/H-2K^b CD8$^+$ T cells

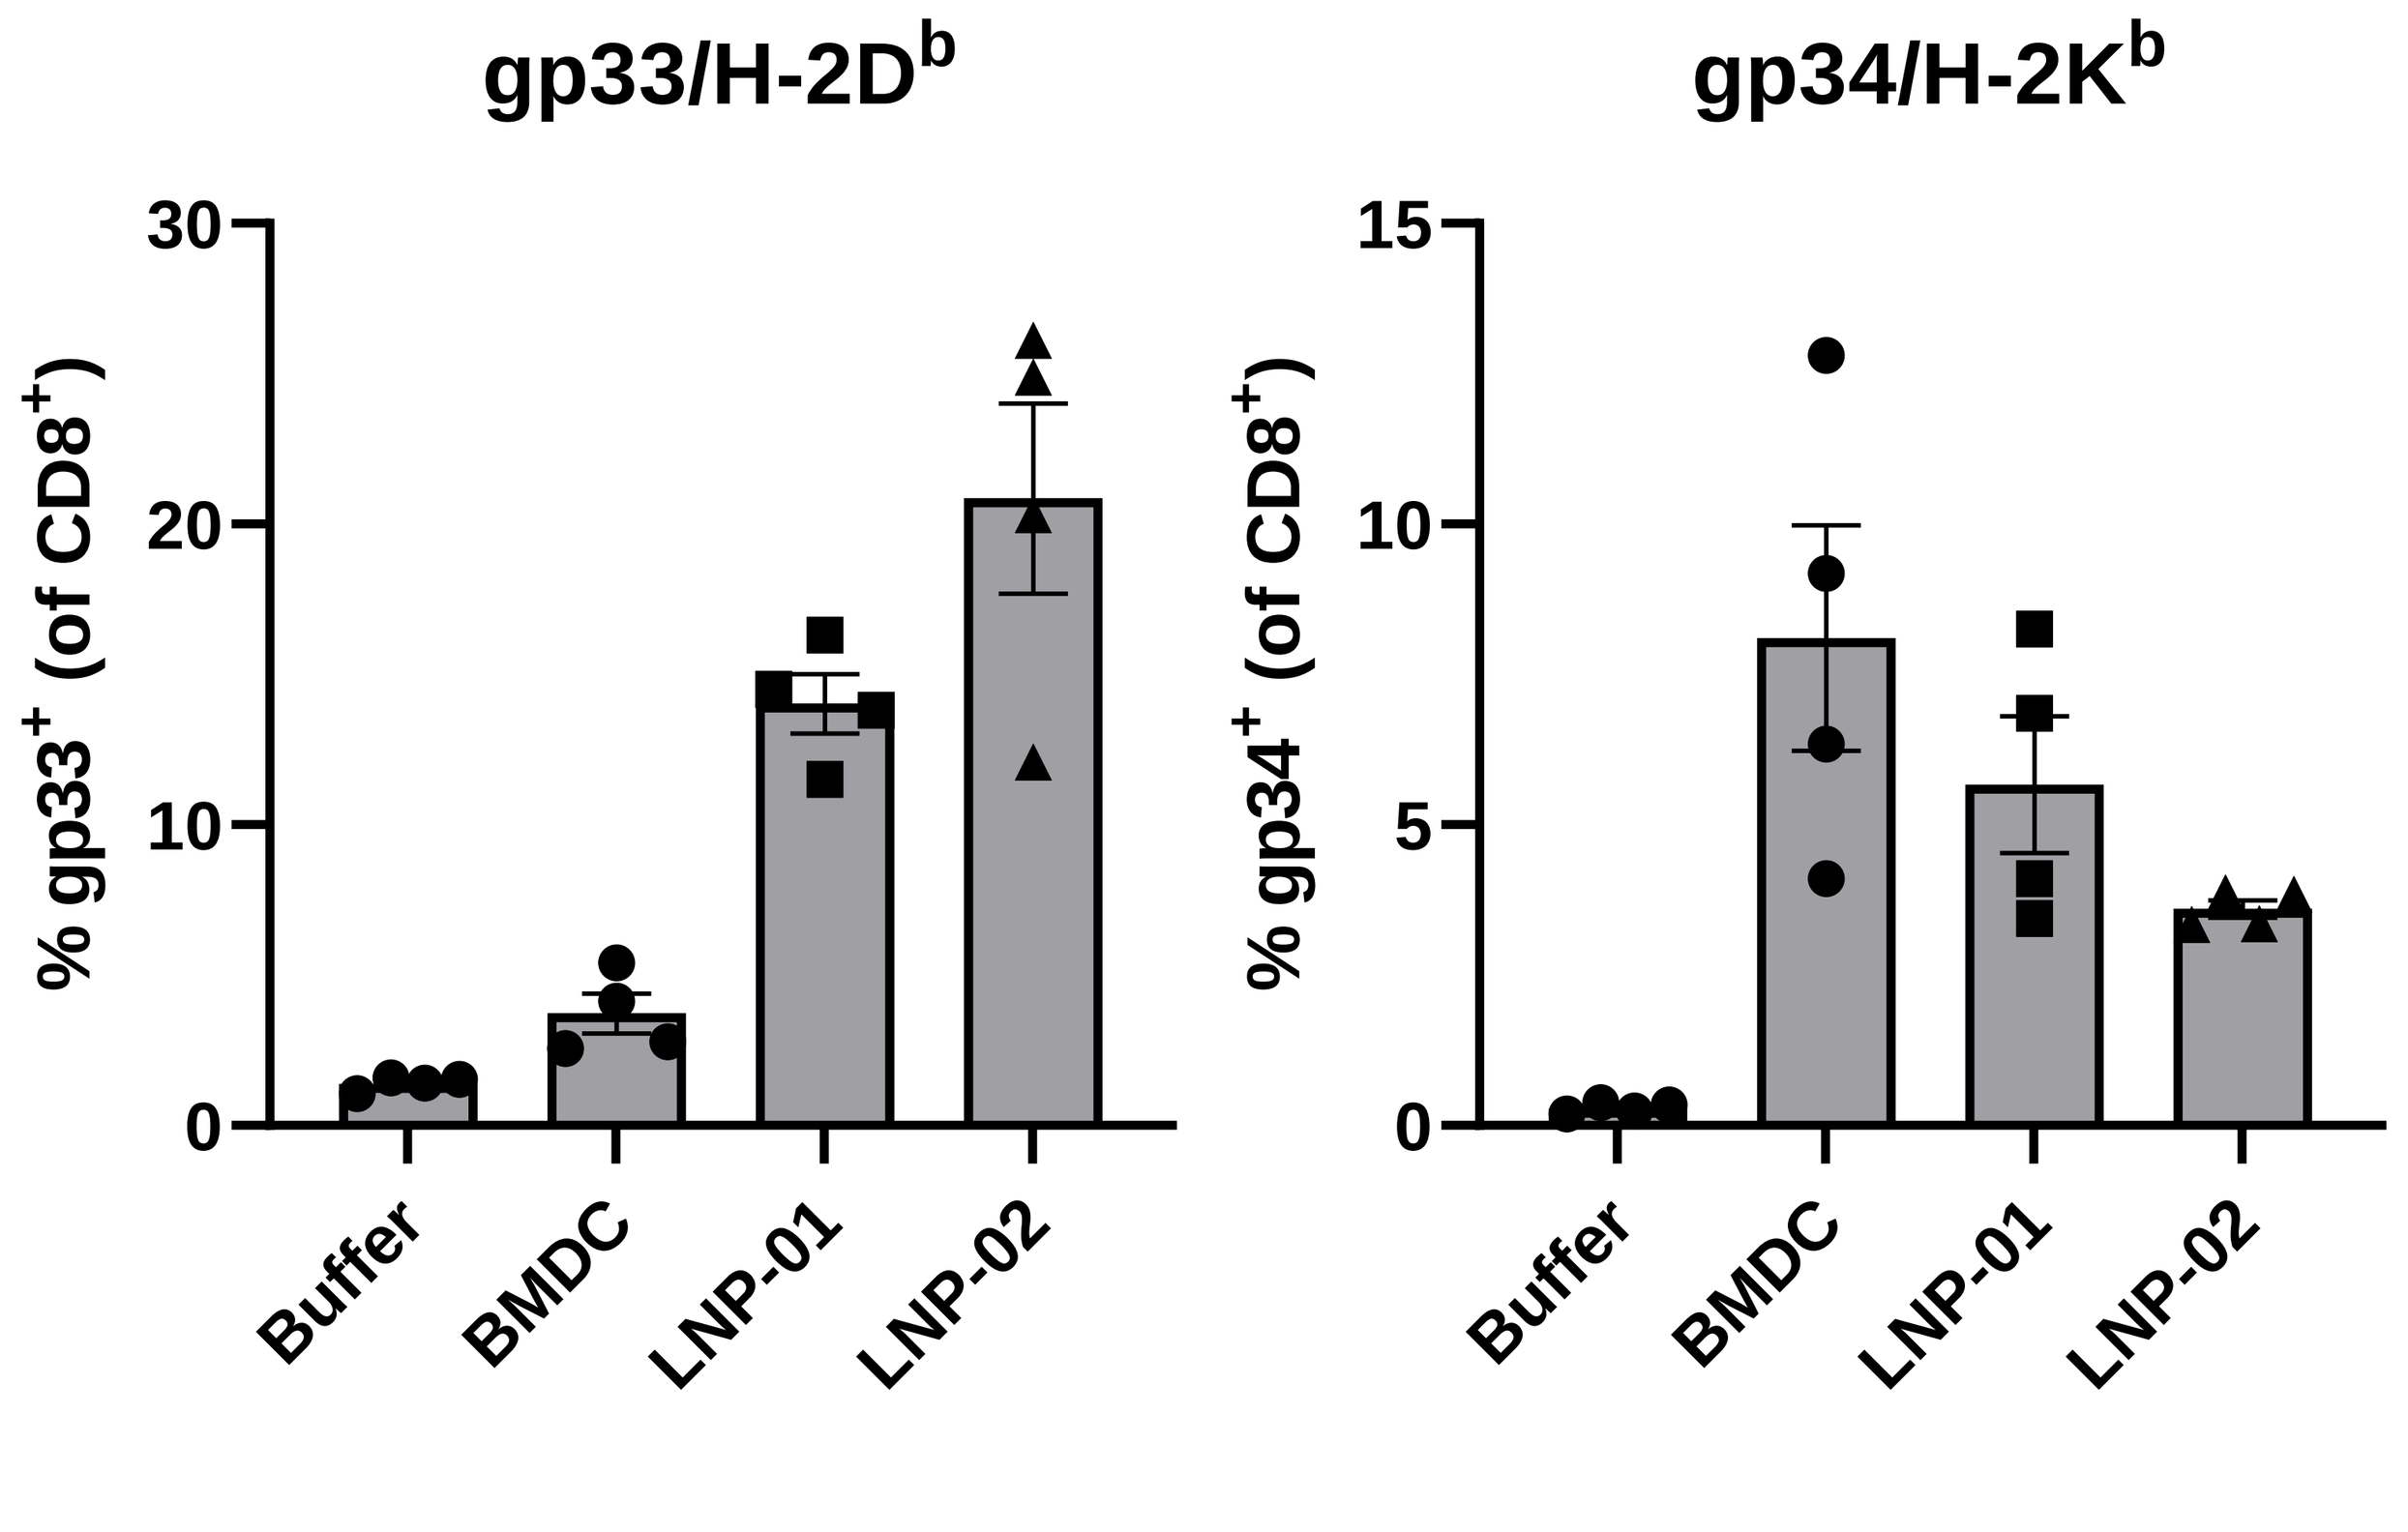
gp33/H-2Db
gp34/H-2Kb
% gp33+ (of CD8+)
% gp34+ (of CD8+)
Buffer
BMDC
LNP-01
LNP-02

Figure 1. LNPs induce strong CD8+ specific equivalent or exceeding those from BMDCs
Female C57BL/6 mice (n=8) were immunized with 50µL of LNPs in the biceps-femoris on day 0 and 4 and peripheral blood was collected on day 8 for tetramer staining. A positive control of bone marrow derived dendritic cells (BMDCs), matured with CpG and pulsed with gp33, gp61 and gp276 was delivered i.p. on days 0, 2 & 4 ($5x10^5$ cells/injection). Peripheral blood was stained for antigen specific CD8$^+$ T cells using tetramers to gp33/H-2D^b and gp34/H-2K^b.

Immunization with next-generation LNP can break immune tolerance for self-antigen in RIPgp mice

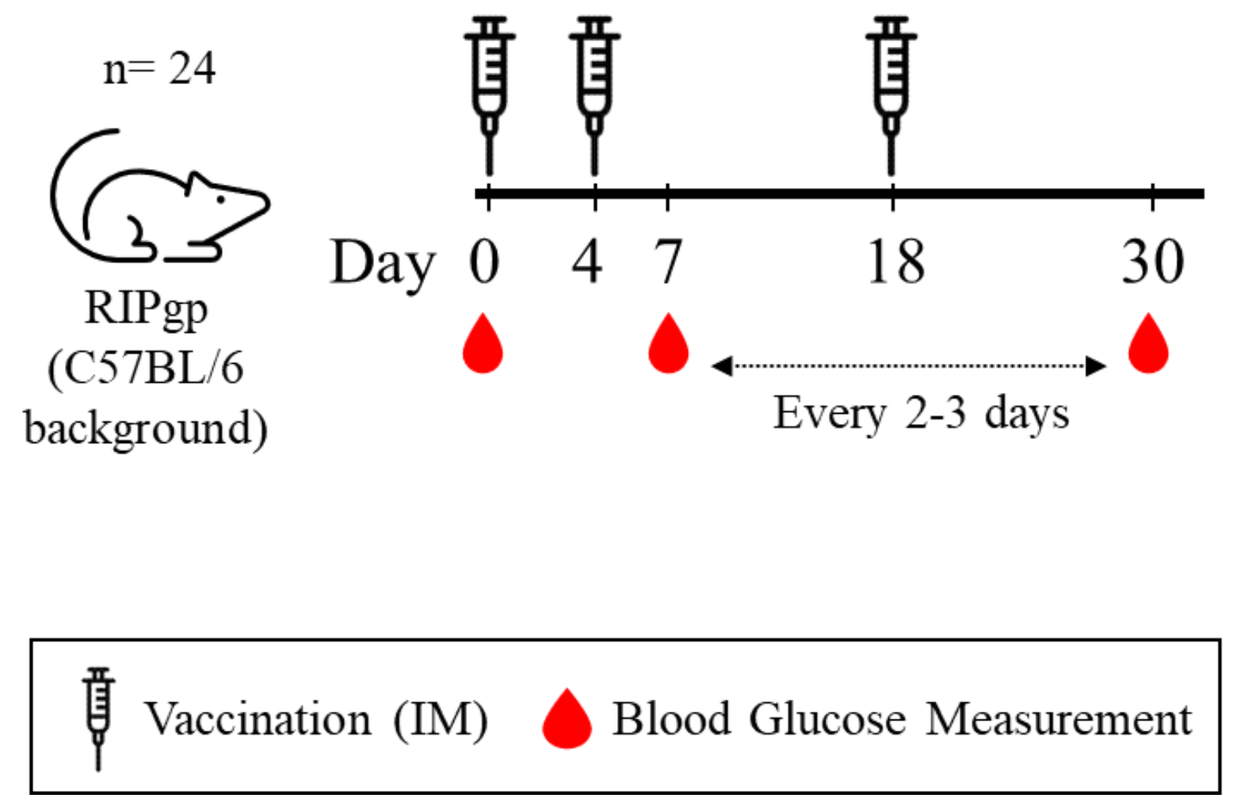
n= 24
RIPgp
(C57BL/6
background)
Day 0 4 7 18 30
Every 2-3 days
Vaccination (IM)
Blood Glucose Measurement

- Despite induction of antigen-specific T cells by LNP-02 (Fig 1), T cells induced by LNP-02 lack the fitness or ability to effectively break self-tolerance
- LNP-01 induces a robust CD8$^+$ T cell response capable of breaking self-tolerance

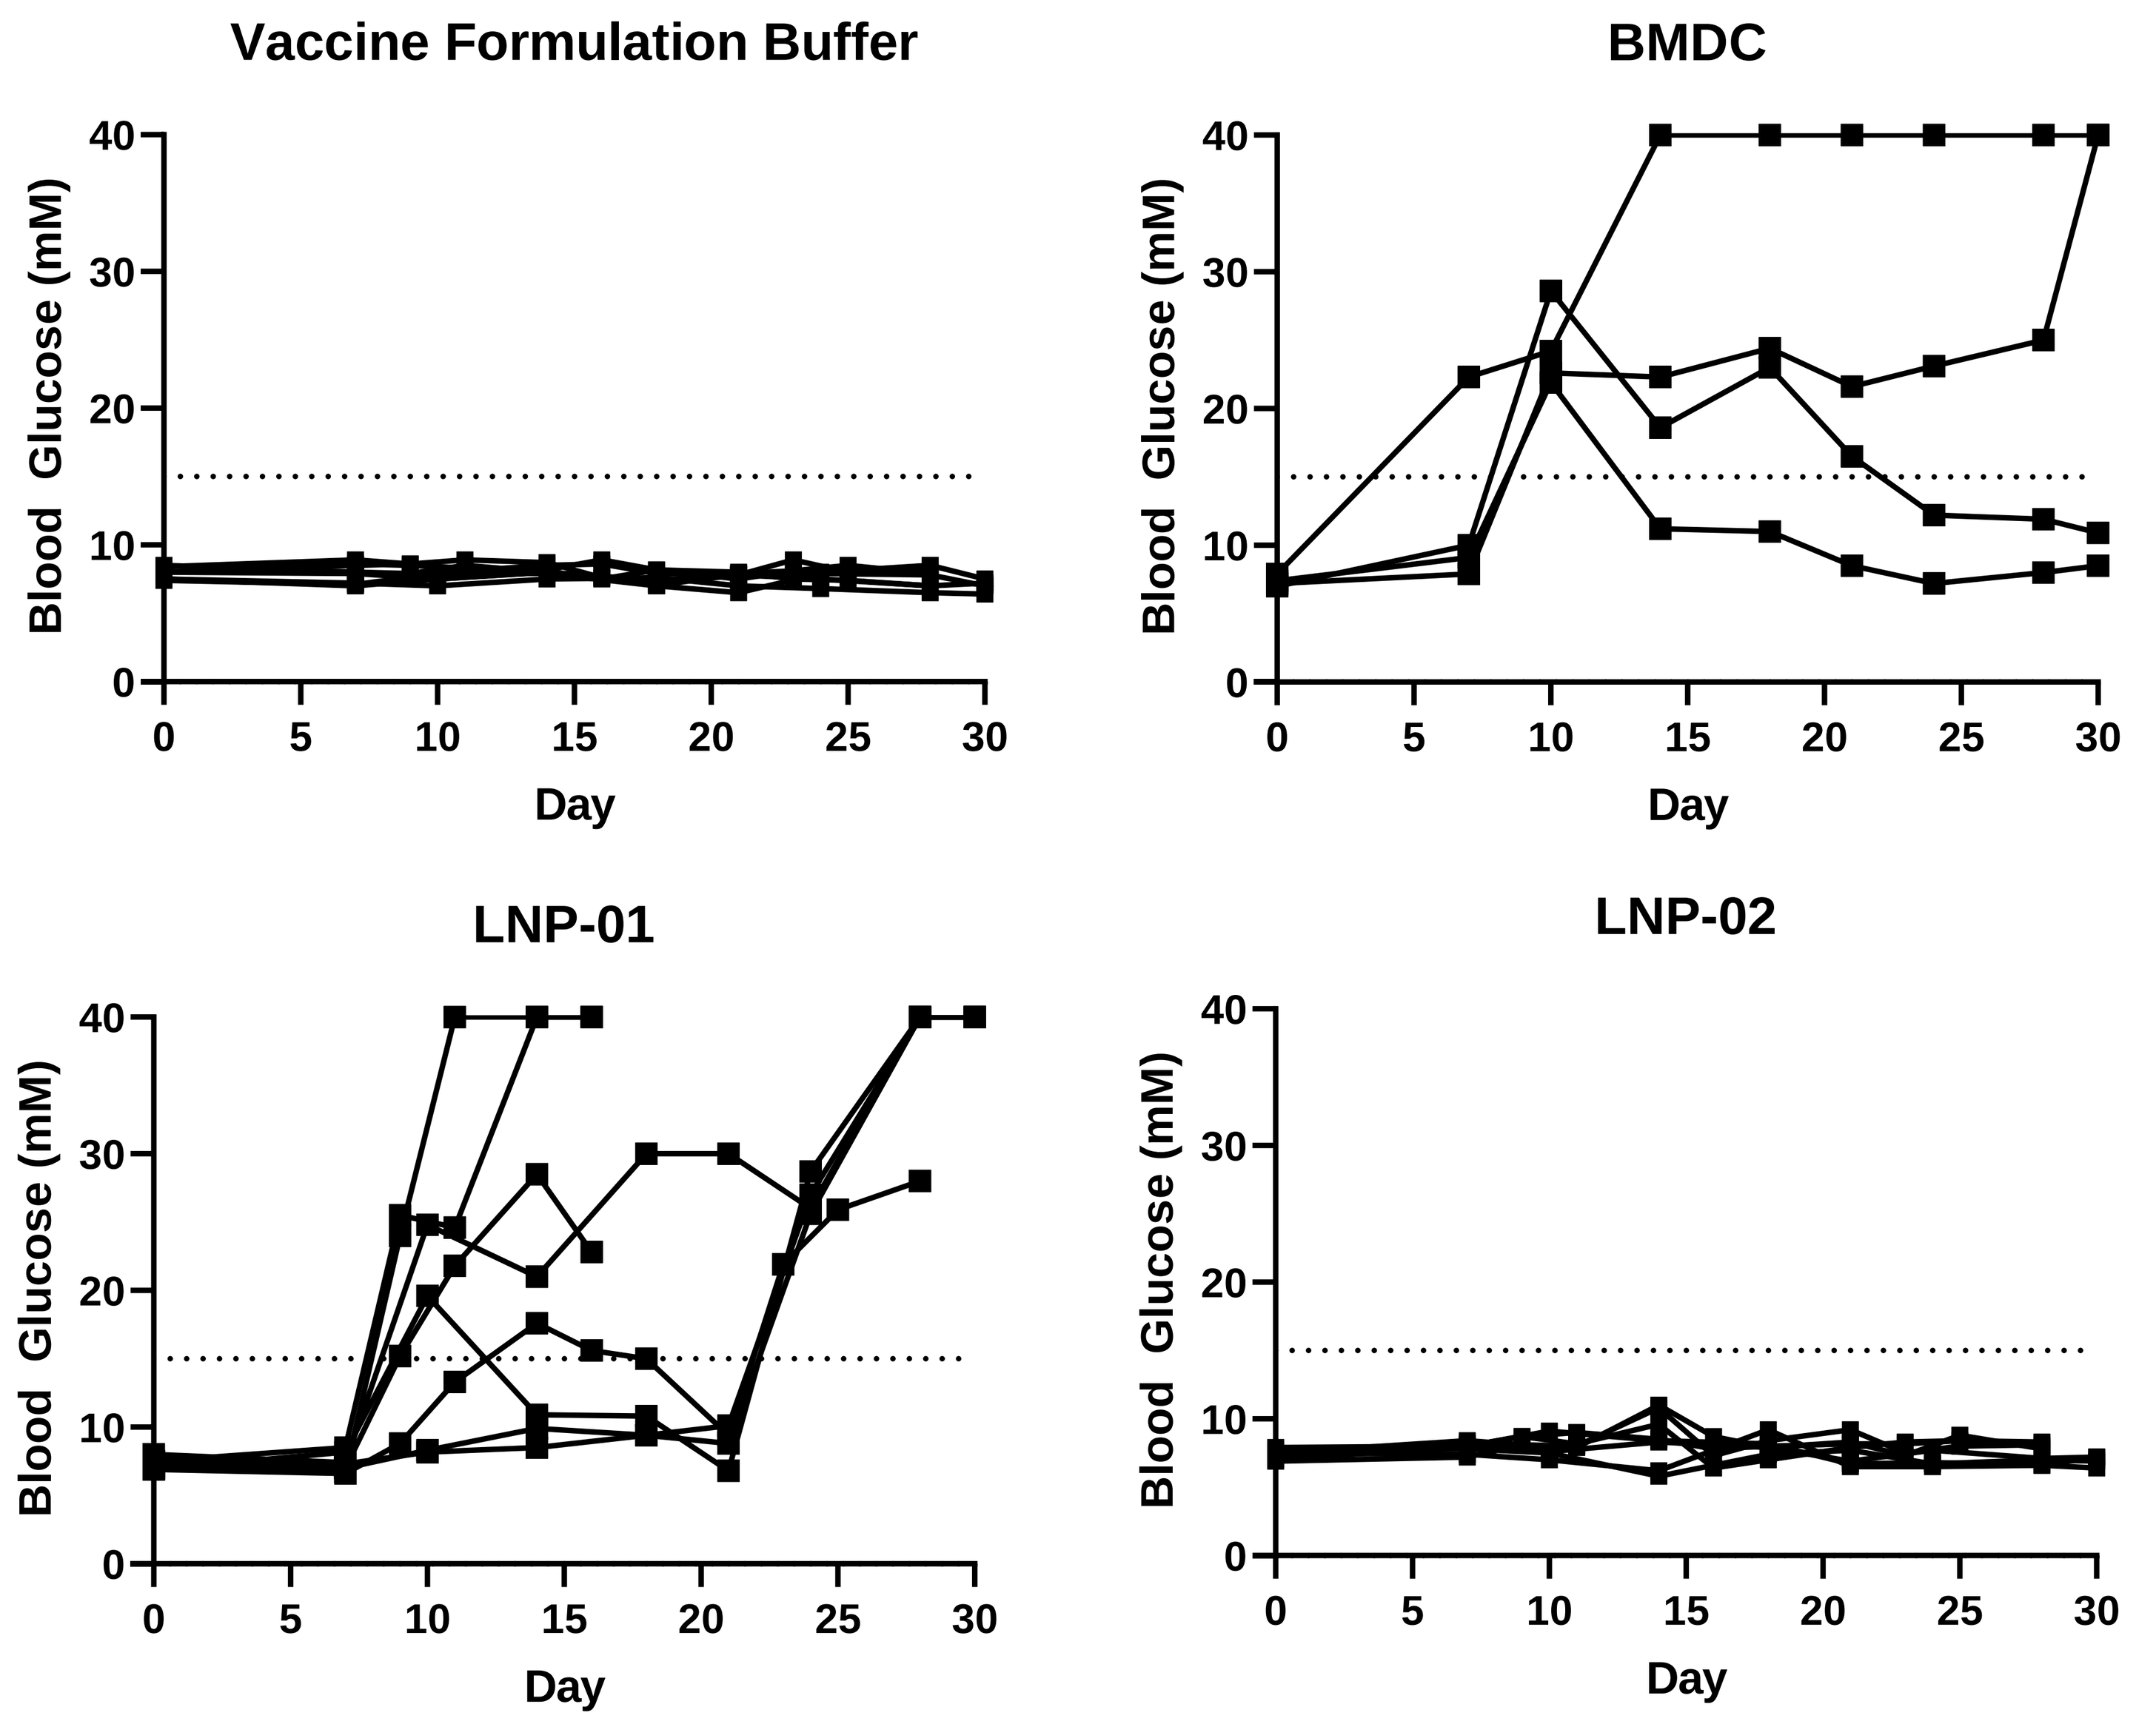
Vaccine Formulation Buffer
BMDC
LNP-01
LNP-02
Blood Glucose (mM)
Day

Figure 2. Next-generation LNP (LNP-01) breaks immune tolerance for self-antigen in RIPgp mice.
RIPgp transgenic mice (n=24) were immunized with 50µL of LNPs in the biceps-femoris on days 0, 4 and 18 and peripheral blood glucose was monitored daily for signs of hyperglycemia (> 15mM). A positive control of bone marrow derived dendritic cells (BMDCs), matured with CpG and pulsed with gp33, gp61 and gp276 was delivered i.p. on days 0, 2 & 4 ($5x10^5$ cells).

Immunization with next-generation LNP inhibits tumor growth and improves survival in MC38gp tumor model

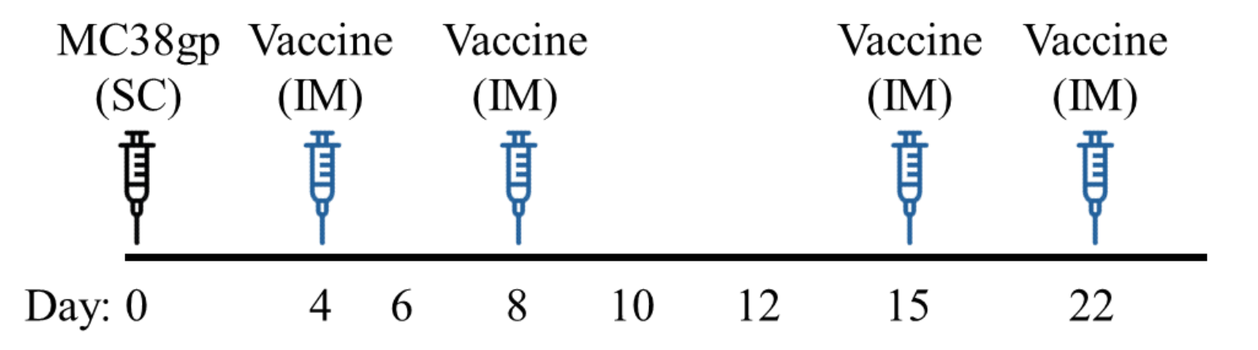
MC38gp (SC)
Vaccine (IM)
Vaccine (IM)
Vaccine (IM)
Vaccine (IM)
Day: 0 4 6 8 10 12 15 22

- LNP-01, showing success in the RIPgp model was able to clear tumor burden in 50% of animals (5/10 CR) as a monotherapy without supplemental CPI

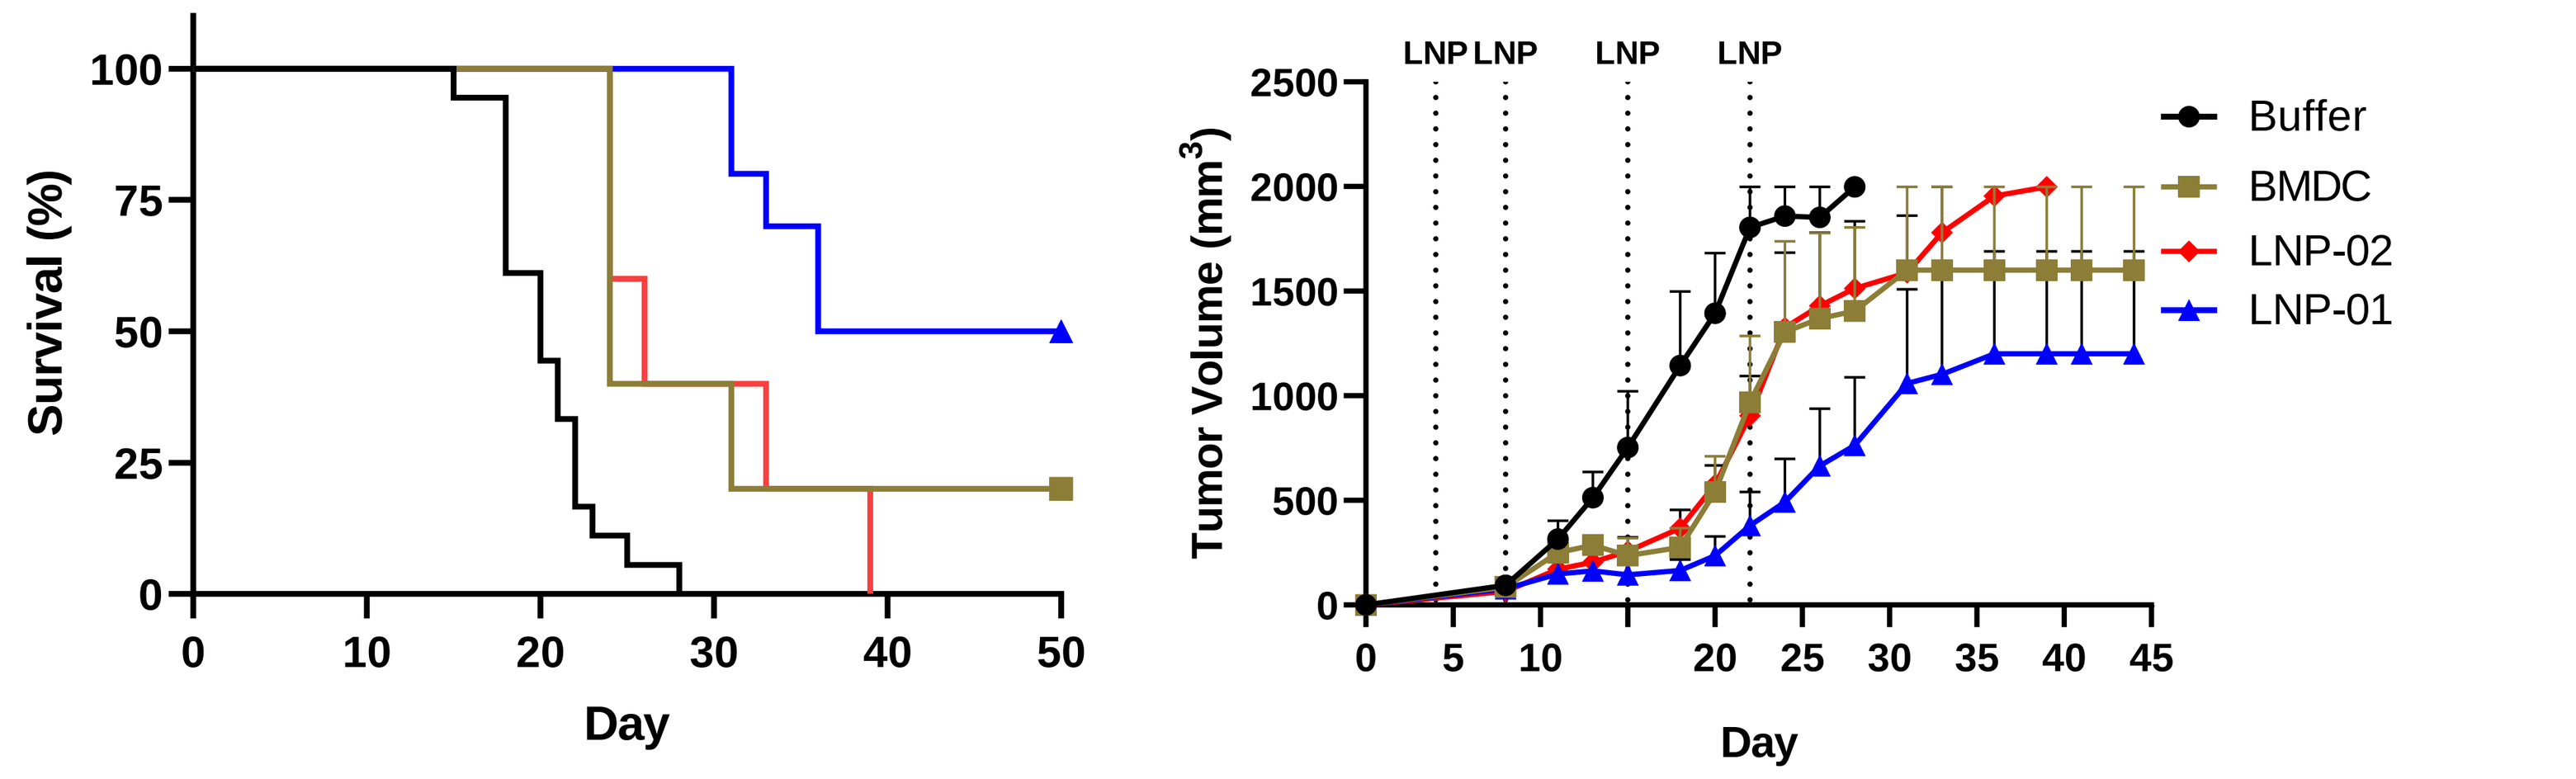
Survival (%)
Day
Tumor Volume (mm3)
LNP LNP LNP LNP
Buffer
BMDC
LNP-02
LNP-01

Figure 3. LNP-01 immunization induces potent anti-tumor immunity to inhibit solid tumor growth and promote survival in MC38gp model
Female C57BL/6 mice were inoculated s.c. with $1x10^6$ MC38gp tumor cells on day 0 and treated with LNP candidates on days 4, 8, 15 & 22 (12.5µg/50µl/dose). BMDCs matured with CpG and pulsed with gp33/61/276 peptides were used as a positive control and delivered i.p. on days 4, 6 and 8 ($5x10^5$ cells/dose). Tumor volume was measured using digital calipers and calculated as Width^2xLength/2. Animals clearing established tumor were deemed to have a complete response (CR).

CONCLUSIONS

- Multiple LNP formulations are effective at generating antigen-specific T cells (Fig 1), however the functional ability of these cells to break self-tolerance and kill target tissue is highly dependent on the LNP formulation
- The RIPgp transgenic model offers a useful screening tool to assess LNPs capable of inducing cytotoxic CD8$^+$ T cells capable of breaking self-tolerance and likely to be efficacious when utilized as an anti-tumor mRNA/LNP based therapeutic treatment
- LNP-01 is capable of breaking self-tolerance and inducing autoimmune diabetes in the RIPgp model as well as offering long-term tumor control when utilized as a therapeutic monotherapy for established MC38gp tumors, as well as in a murine model of glioblastoma (GBM, Abstract # 5002)
- Additional POC for LNP-01 formulated mRNA cancer vaccines for multiple solid tumors are ongoing as Providence Therapeutic's program moves towards clinical trials in 2025

CONTACT

Business Development: bd@providencetherapeutics.com
Investor Relations: investor@providencetherapeutics.com